Anemia in Long-Term Care Patients: Prevalence and Relationship to Falls

Tommy Philpot, PharmD[1], Brahim Bookhart, MBA, MPH[2], Victor Lampasona, PharmD[2], Samir H. Mody, PharmD, MBA[2]
[1]Profiles and Interventions, Inc., Atlanta, GA; [2]Ortho Biotech Clinical Affairs, LLC, Bridgewater, NJ

OBJECTIVE

- To quantify the prevalence of anemia in the long-term care (LTC) setting and to explore the association between anemia and falls in LTC patients.

BACKGROUND

- The incidence of falls in nursing homes is high, with 1.5 falls occurring per bed per year.[1]
- Thirty percent of the community-dwelling elderly fall annually and this risk increases to 50% by the age of 80. Serious injuries caused by a fall, such as fractures and head injuries, are sustained by 10% of the elderly and often lead to functional disability, increased health care costs, and increased mortality.[2]
- The most important underlying risk factors for falls and injuries include comorbid conditions, lower-extremity weakness, gait and balance instability, poor vision, cognitive and functional impairment, and sedating and psychoactive medications.[1]
- Higher risk for falls also may be associated with anemia in nursing home patients. In one study of older adults (from nursing homes and the community) hospitalized for hip fracture, anemia was associated with a nearly threefold adjusted increased risk of falls (p=0.041).[3] Anemic residents who suffer a hip fracture subsequent to falling were found to be at higher risk for increased mortality.[4]
- In addition, anemia in patients with chronic kidney disease (CKD) has been found to be a significant predictor of mortality. Recent data from Collins[5] et al. revealed that patients with chronic kidney disease (CKD), when combined with anemia, have a mortality risk 3.7 times higher than those without either condition.

METHODS

RESIDENTS AND STUDY DESIGN

- Data were obtained from a sample of residents of 26 LTC facilities based in North Carolina, Georgia, and Pennsylvania who met the following inclusion criteria:
 - ≥65 years of age
 - Resided in the facility for ^{3}6 months
 - Not receiving renal dialysis
 - Had the following information available in their record:
 - MDS assessment
 - Hemoglobin (Hb) and serum creatinine values (6 or fewer months prior to the most recent MDS assessment)
 - Race
 - Gender
- Data collected for time period of February 2005 through September 2006
- The date of the most recent MDS assessment was identified. All falling episodes within 180 days of the MDS assessment date were identified.
- Anemia status was defined using WHO criteria assessment (Hb <12 g/dL for women; <13 g/dL for men) for the most recent Hb level prior to the MDS assessment.
- All data were de-identified in accordance with HIPAA rules prior to analysis.
- All incidents of falls were obtained from the MDS assessment.
- An odds ratio (OR) calculated the association between anemia status and falls.

RESULTS

PATIENT CHARACTERISTICS

- 804 patients met the inclusion criteria.
- Mean age was 83 years; 81% of patients were female.
- 52% of all patients studied were anemic, with 55% of anemic patients having a Hb level ≤11 g/dL.

	Total	Anemia Status		
	All	*Not Anemic*	*Anemic*	*p-value**
	n	***n***	***n***	
Total Patients	804	386	418	
	% or mean (sd)	*% or mean (sd)*	*% or mean (sd)*	
Age	85 (7.8)	84.6 (7.7)	85.2 (7.9)	NS
Female	81%	83%	79%	NS
African-American	21%	13%	29%	<0.001
Hemoglobin Level (g/dL)	12.0 (1.6)	13.3 (0.97)	10.8 (0.97)	<0.001
GFR [MDRD], mL/min/1.73 m^2	71.7 (24.8)	74.5 (23.5)	69.2 (25.7)	0.002
Low GFR, <60 mL/min/1.73 m^2	35%	29%	41%	0.001
Received an Erythropoietic-Stimulating Agent	2%	1%	3%	0.011

Terms: GFR, glomerular filtration rate; MDRD, Modification of Diet in Renal Disease; WHO definition of anemia = index Hb <12 g/dL if female, <13 g/dL if male
* t-tests used for differences in means; Chi-square tests used for proportions.

ANEMIA, GENDER AND RENAL STAGE

- Rates of anemia were higher for males (58% anemic) than females (51% anemic) though mean Hb level was 0.7 g/dL lower for females than males.
- A decline in renal function below 60 mL/min/1.73 m2 was associated with a progressive decline in Hb level and an increase in the rate of anemia.

Renal Stage*‡	Male				Female				Total			
	Hemoglobin			*Anemic*	*Hemoglobin*			*Anemic*	*Hemoglobin*			*Anemic*
	n	*Mean (g/dL)*	*SD*	*%*	*n*	*Mean (g/dL)*	*SD*	*%*	*n*	*Mean (g/dL)*	*SD*	*%*
All Stages	151	12.6	1.8	58%	653	11.9	1.5	51%	804	12.0	1.6	52%
Stage 1 CKD	44	12.9	1.5	48%	99	12.0	1.5	47%	143	12.3	1.6	48%
Stage 2 CKD	74	12.6	1.8	58%	302	12.1	1.5	45%	376	12.2	1.5	47%
Stage 3 CKD	30	12.0	1.8	67%	230	11.7	1.5	58%	260	11.7	1.6	59%
Stage 4 CKD	3	11.3	0.5	100%	17	11.0	1.6	71%	20	11.0	1.5	75%
Stage 5 CKD	0	n/a	n/a	n/a	1	13.6	n/a	100%	1	13.6	n/a	100%

Terms: CKD, chronic kidney disease; WHO definition of anemia = index Hb <12 g/dL if female, <13 g/dL if male
* Stage 1 = GFR of 90 or higher; Stage 2 = 60 to 89; Stage 3 = 30 to 59; Stage 4 = 15 to 29; Stage 5 = 15 or lower
‡ 4 patients were missing a GFR value and could not be assigned to a specific stage

RATES OF FALLS AND ANEMIA

- At least one falling episode was recorded for 23% of the study population.
- A higher proportion of anemic patients fell compared to non-anemic patients, 27% vs. 18%, respectively.
- Anemic patients had a 68% higher likelihood of falling compared to non-anemics (OR: 1.68; 95% Confidence Interval: 1.20, 2.36).

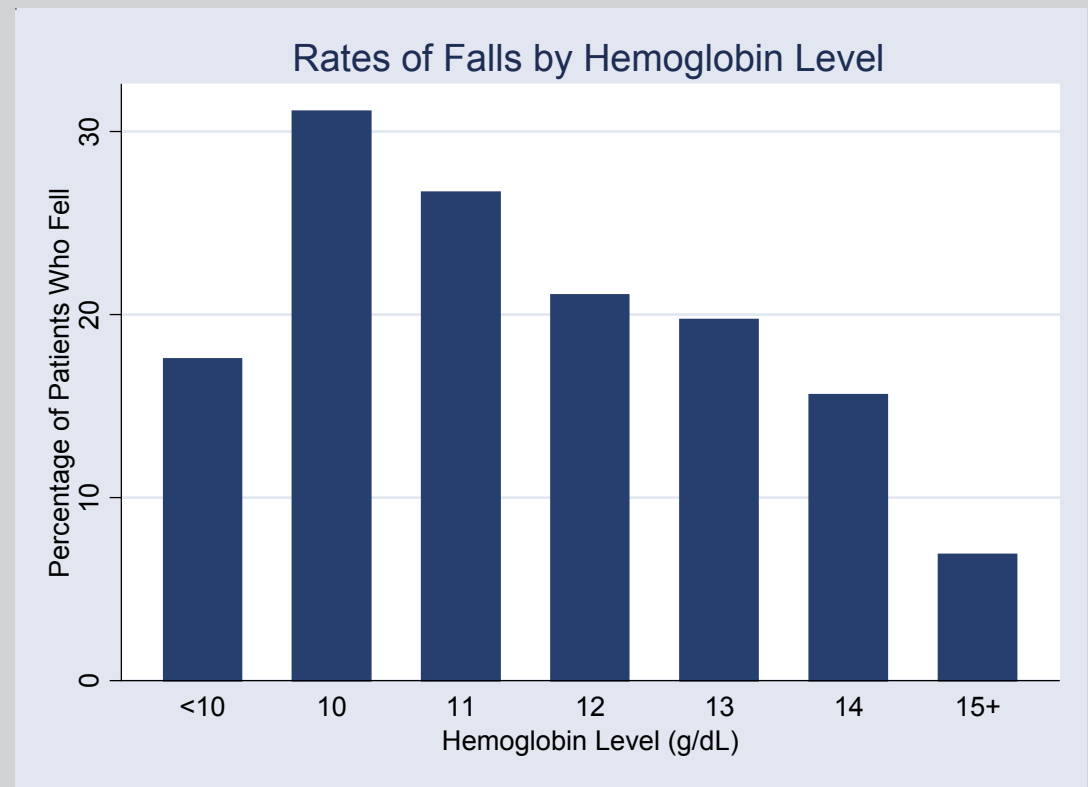
Rates of Falls by Hemoglobin Level
Percentage of Patients Who Fell
0
10
20
30
<10
10
11
12
13
14
15+
Hemoglobin Level (g/dL)

ASSOCIATION BETWEEN ANEMIA AND FALLS

- Of the variables examined, only anemia status (62% vs. 49%, p=0.002) and mean Hb level (11.8 g/dL vs. 12.1 g/dL, p=0.005) were significantly different between the cohort of patients who fell vs. those who did not fall.

	Total	Falling Status		
	All	*Did Not Fall*	*Fell*	*p-value**
	n	***n***	***n***	
Total Patients	804	623	181	
	% or mean (sd)	*% or mean (sd)*	*% or mean (sd)*	
Anemic	52%	49%	62%	0.002
Age	85.0 (7.8)	84.8 (7.8)	85.3 (7.8)	NS
Female	81%	82%	80%	NS
African-American	21%	22%	18%	NS
Hemoglobin Level (g/dL)	12.0 (1.6)	12.1 (1.6)	11.8 (1.4)	0.005
GFR [MDRD], mL/min/1.73 m^2	71.7 (24.8)	72.4 (25.3)	69.5 (23.0)	NS
Low GFR, <60 mL/min/1.73 m^2	35%	34%	40%	NS
Received an Erythropoietic-Stimulating Agent	2%	2%	3%	NS

Terms: GFR, glomerular filtration rate; MDRD, Modification of Diet in Renal Disease; WHO definition of anemia = index Hb <12 g/dL if female, <13 g/dL if male
* t-tests used for differences in means; Chi-square tests used for proportions.

LIMITATIONS

- Study limitations include
 - the retrospective/cross-sectional study design
 - non-random selection of nursing homes
 - the retrospective/cross-sectional study design
 - potential confounding of additional covariates not collected
 - potential incompleteness of chart data

CONCLUSIONS

- In this large multi-state study of LTC patients, the majority of patients had anemia.
- Anemia appears to progress substantially with a decline on GFR below 60 mL/min/1.73 m^2 (Stage 3 or higher).
- Finally, anemia was associated with a significantly higher risk of falling.
 - The results of the relationship between anemia and falls were consistent with published data on community dwelling elderly patients.[3]
- Since falls, and related consequences such as fractures and hospital visits, are associated with significant morbidity and mortality, anemia should be actively monitored in LTC patients.

REFERENCES

[1] [Rubenstein 1994]
[2] [Duh 2005]
[3] Dharmarajan TS, Norkus EP. J Am Med Dir Assoc 2004;5:395-400.
[4] Gruson KI, Aharonoff GB, Egol KA, et al. J Orthop Trauma. 2002;16(1):39-44.
[5] [Collins Adv Stud med. 2003;3:S194-S197]
[6] [Silverberg DS, et al. Nephrol Dial Transplant. 2003;18:141-146.]

Presented at the International Society for Pharmacoeconomic and Pharmaceutical Outcomes Research (ISPOR) Annual Meeting, May 19-22, 2007, Arlington, VA, USA.

This research was supported by Ortho Biotech Clinical Affairs, LLC, Bridgewater, NJ, USA.